HIGH-THROUGHPUT APPLICATIONS FOR SCREENING GPCR ACTIVATION USING bioSensAll®

Frank Cao[1], Claire Normand[1], Arturo Mancini[1], Samya Aouad[1], Herthana Kandasamy[1], Robert Cerone[1], Meriem Semache[1], Melissa R. Miller[2], Shunjie Guan[2], Dong Yan[2], Craig L. Hyde[2], Jean-Philippe Fortin[2], Eric B. Fauman[2], Stephan Schann[1], and Laurent Sabbagh[1]

[1]Domain Therapeutics, Montreal, Quebec, Canada, and Strasbourg, France and [2] Worldwide Research, Development and Medical, Pfizer Global R&D, Cambridge, MA, USA

Abstract

G protein-coupled receptors (GPCRs) represent the largest class of membrane-bound proteins which regulate physiological processes by engaging diverse signaling pathways. Although dysregulation of GPCR activity has been linked to pathological consequences, the involvement of specific signaling pathways toward distinct clinical repercussions remain unresolved. Thus, interrogating the complex signaling behaviors of GPCRs is not only important for understanding GPCR biology, but also essential for successfully targeting these receptors as therapeutic avenues. Here, we describe the application of our bioSens-All® technology within the context of a high throughput screening platform as well as a mechanical stimulation approach to characterize GPCR signaling.

bioSens-All® consists of a panel of 16 selective enhanced bystander bioluminescence resonance energy transfer (ebBRET)-based biosensors that monitor the activation of heterotrimeric G proteins and β-arrestins upon GPCR activation. To achieve high-throughput volumes, bioSens-All® assays were miniaturized and adapted to a 384-well format, where HEK293 cells were co-transfected with receptors and biosensors. Assays were performed using a laboratory automation workflow software to coordinate the interaction between various instruments via a robotic microplate mover. Through this automated platform, we assessed signaling profiles of ~12,000 clinically relevant variants across 60 GPCRs. Analysis was performed through an automated tool to generate and compile over 120,000 concentration-response curves. Signaling profiles for a subset of variants revealed gain- or loss-of-function properties across different signaling pathways thus also allowing us to identify genetic alterations that impart biased signaling profiles for each profiled GPCR.

Using the bioSens-All® technology in a separate application, we developed a novel *in vitro* assay detecting adhesion GPCR ADGRE5 activation following mechanical stimulation. In this assay, cells co-transfected with receptors and biosensors were seeded in 96-well plates and subjected to vigorous orbital shaking prior to BRET measurements. We demonstrated that mechanostimulation of full-length human ADGRE5-transfected cells resulted in a rapid time-dependent recruitment of β-arr2 to the plasma membrane, while such mechanosensitivity was not observed with a control receptor (mu opioid receptor, MOR) despite MOR's ability to engage β-arr2 following ligand stimulation. Finally, the assay demonstrated a Z' value of 0.73, thus highlighting its compatibility for high-throughput screening of novel therapeutic agents acting to modulate physiologically relevant modes of ADGRE5 activation.

Altogether, these applications of our bioSens-All® technology allows interrogation of diverse activation mechanisms and complex signaling behaviors of GPCRs. Ultimately, such approaches can be applied to other GPCRs to facilitate the validation of novel therapeutic targets and discovery of innovative therapeutic agents.

Instrumentation

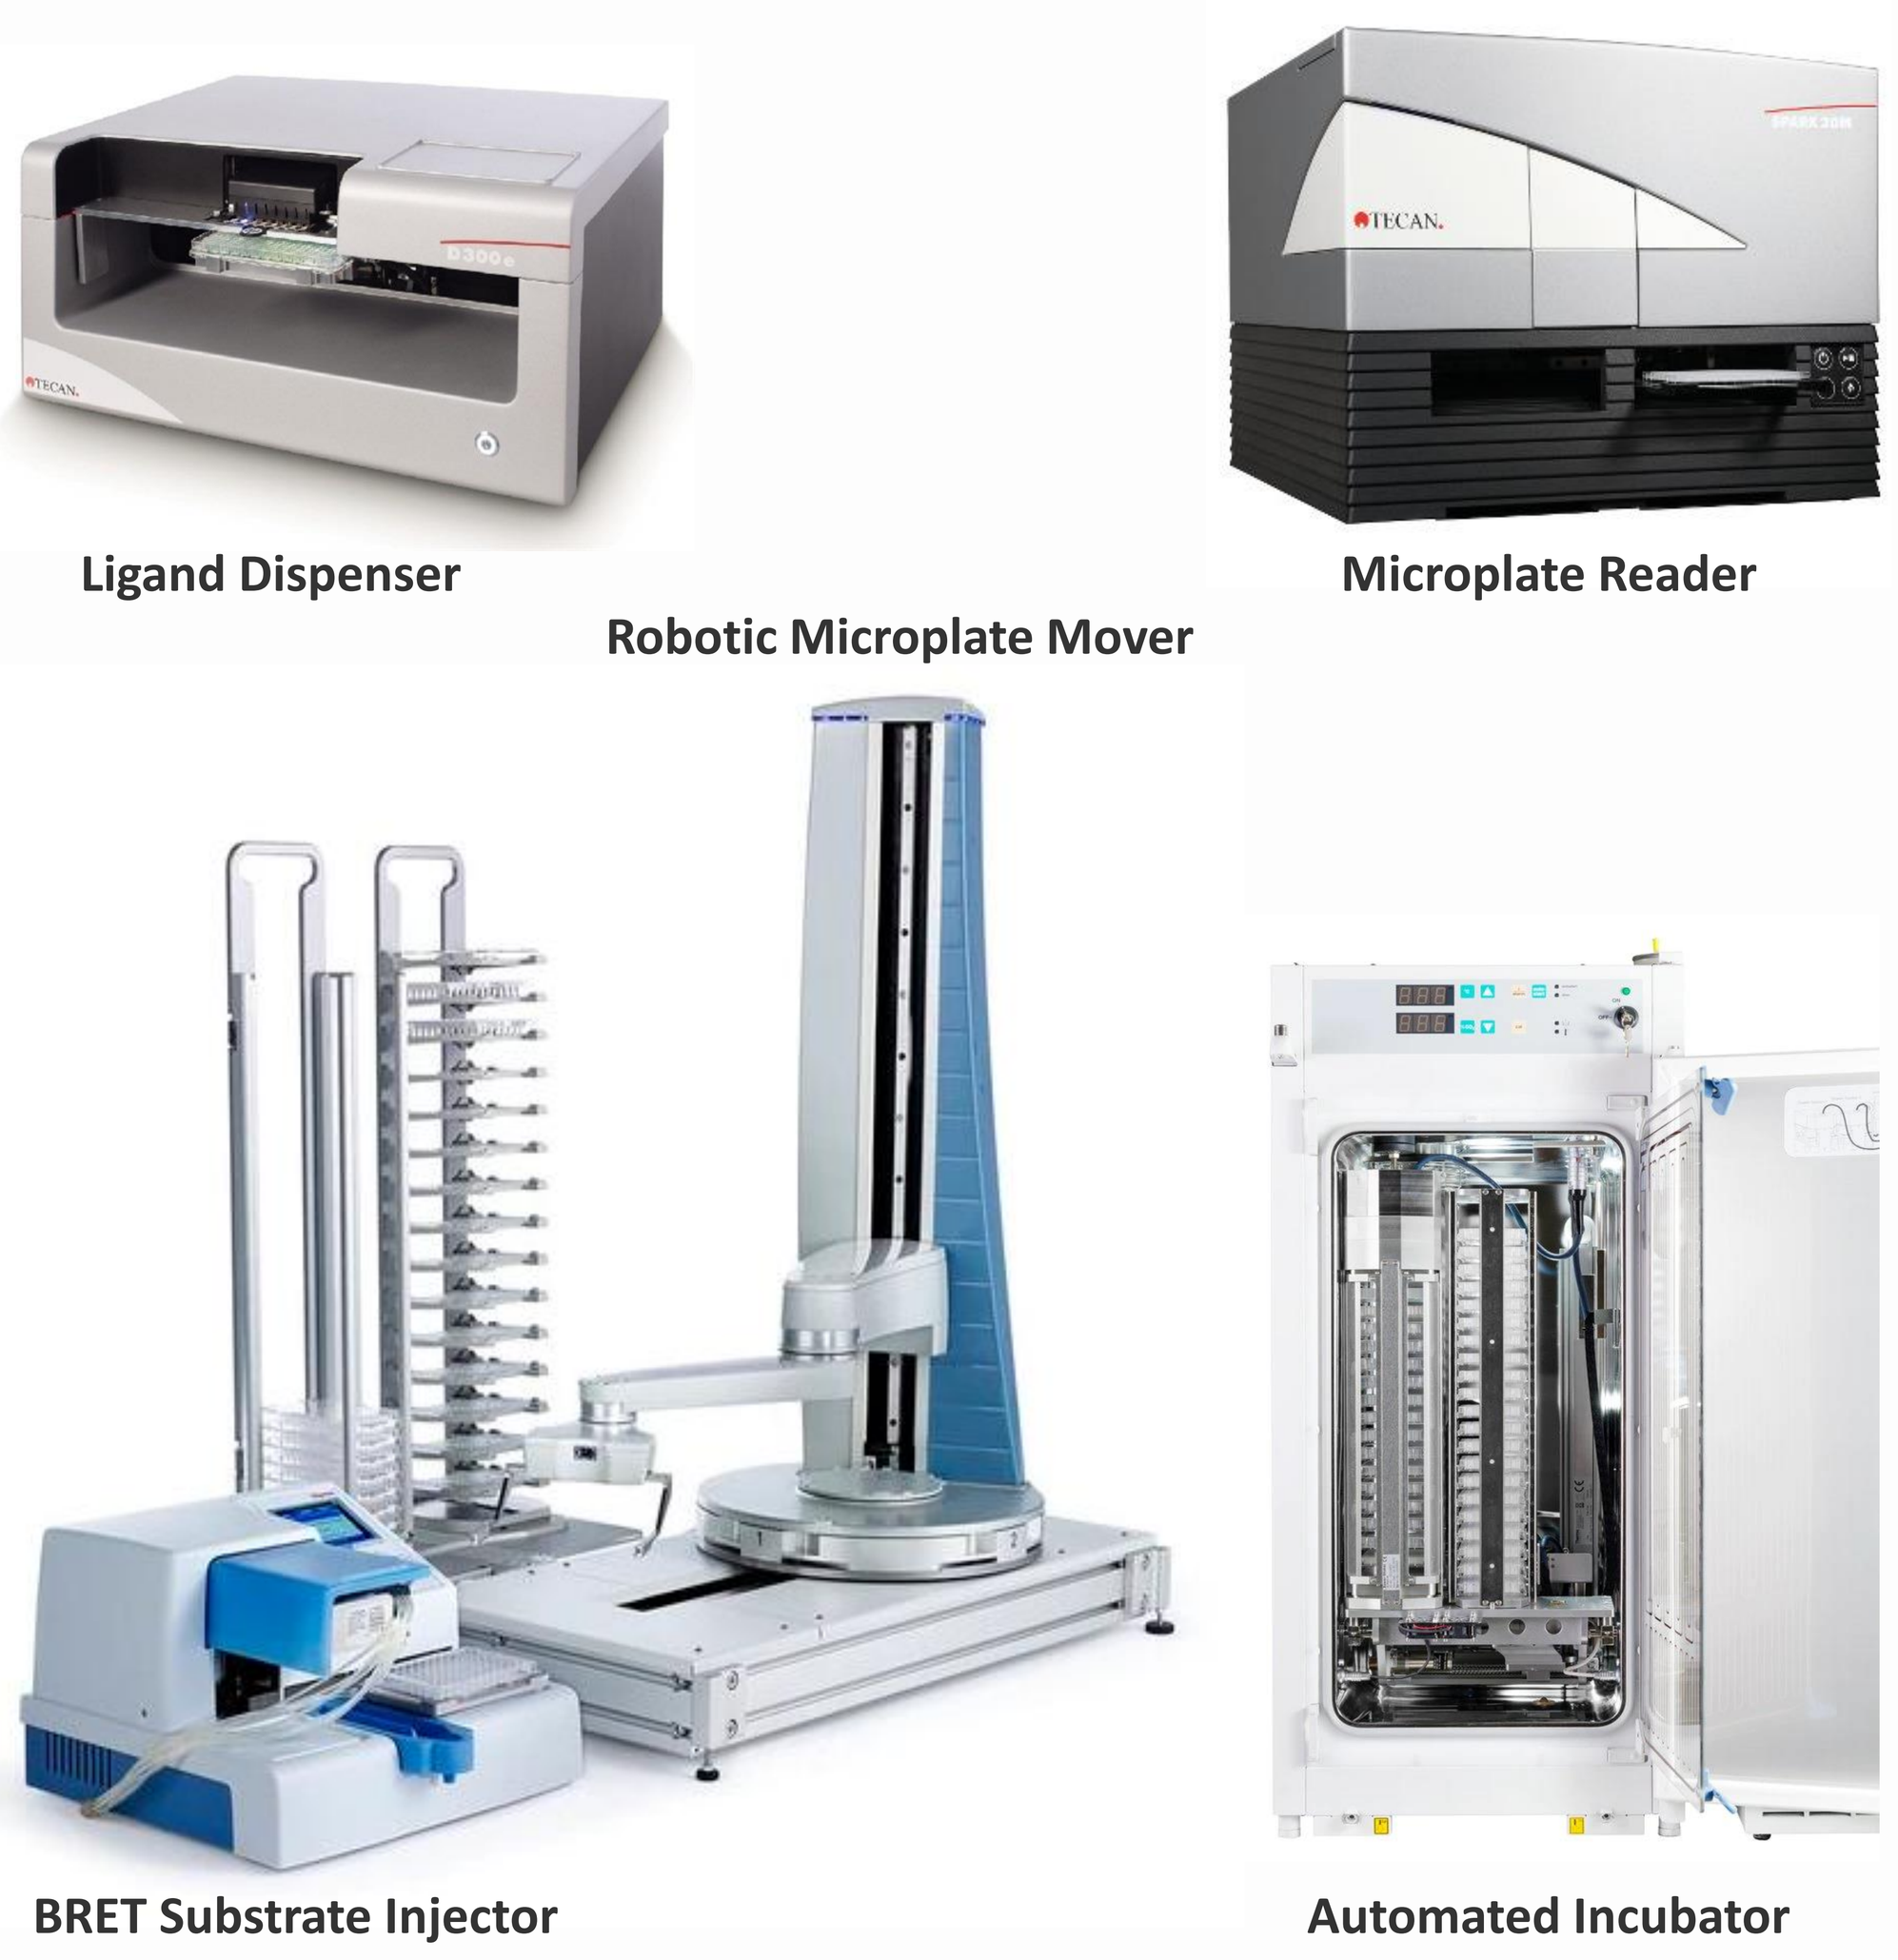
Ligand Dispenser
Robotic Microplate Mover
Microplate Reader
BRET Substrate Injector
Automated Incubator

bioSens-All®

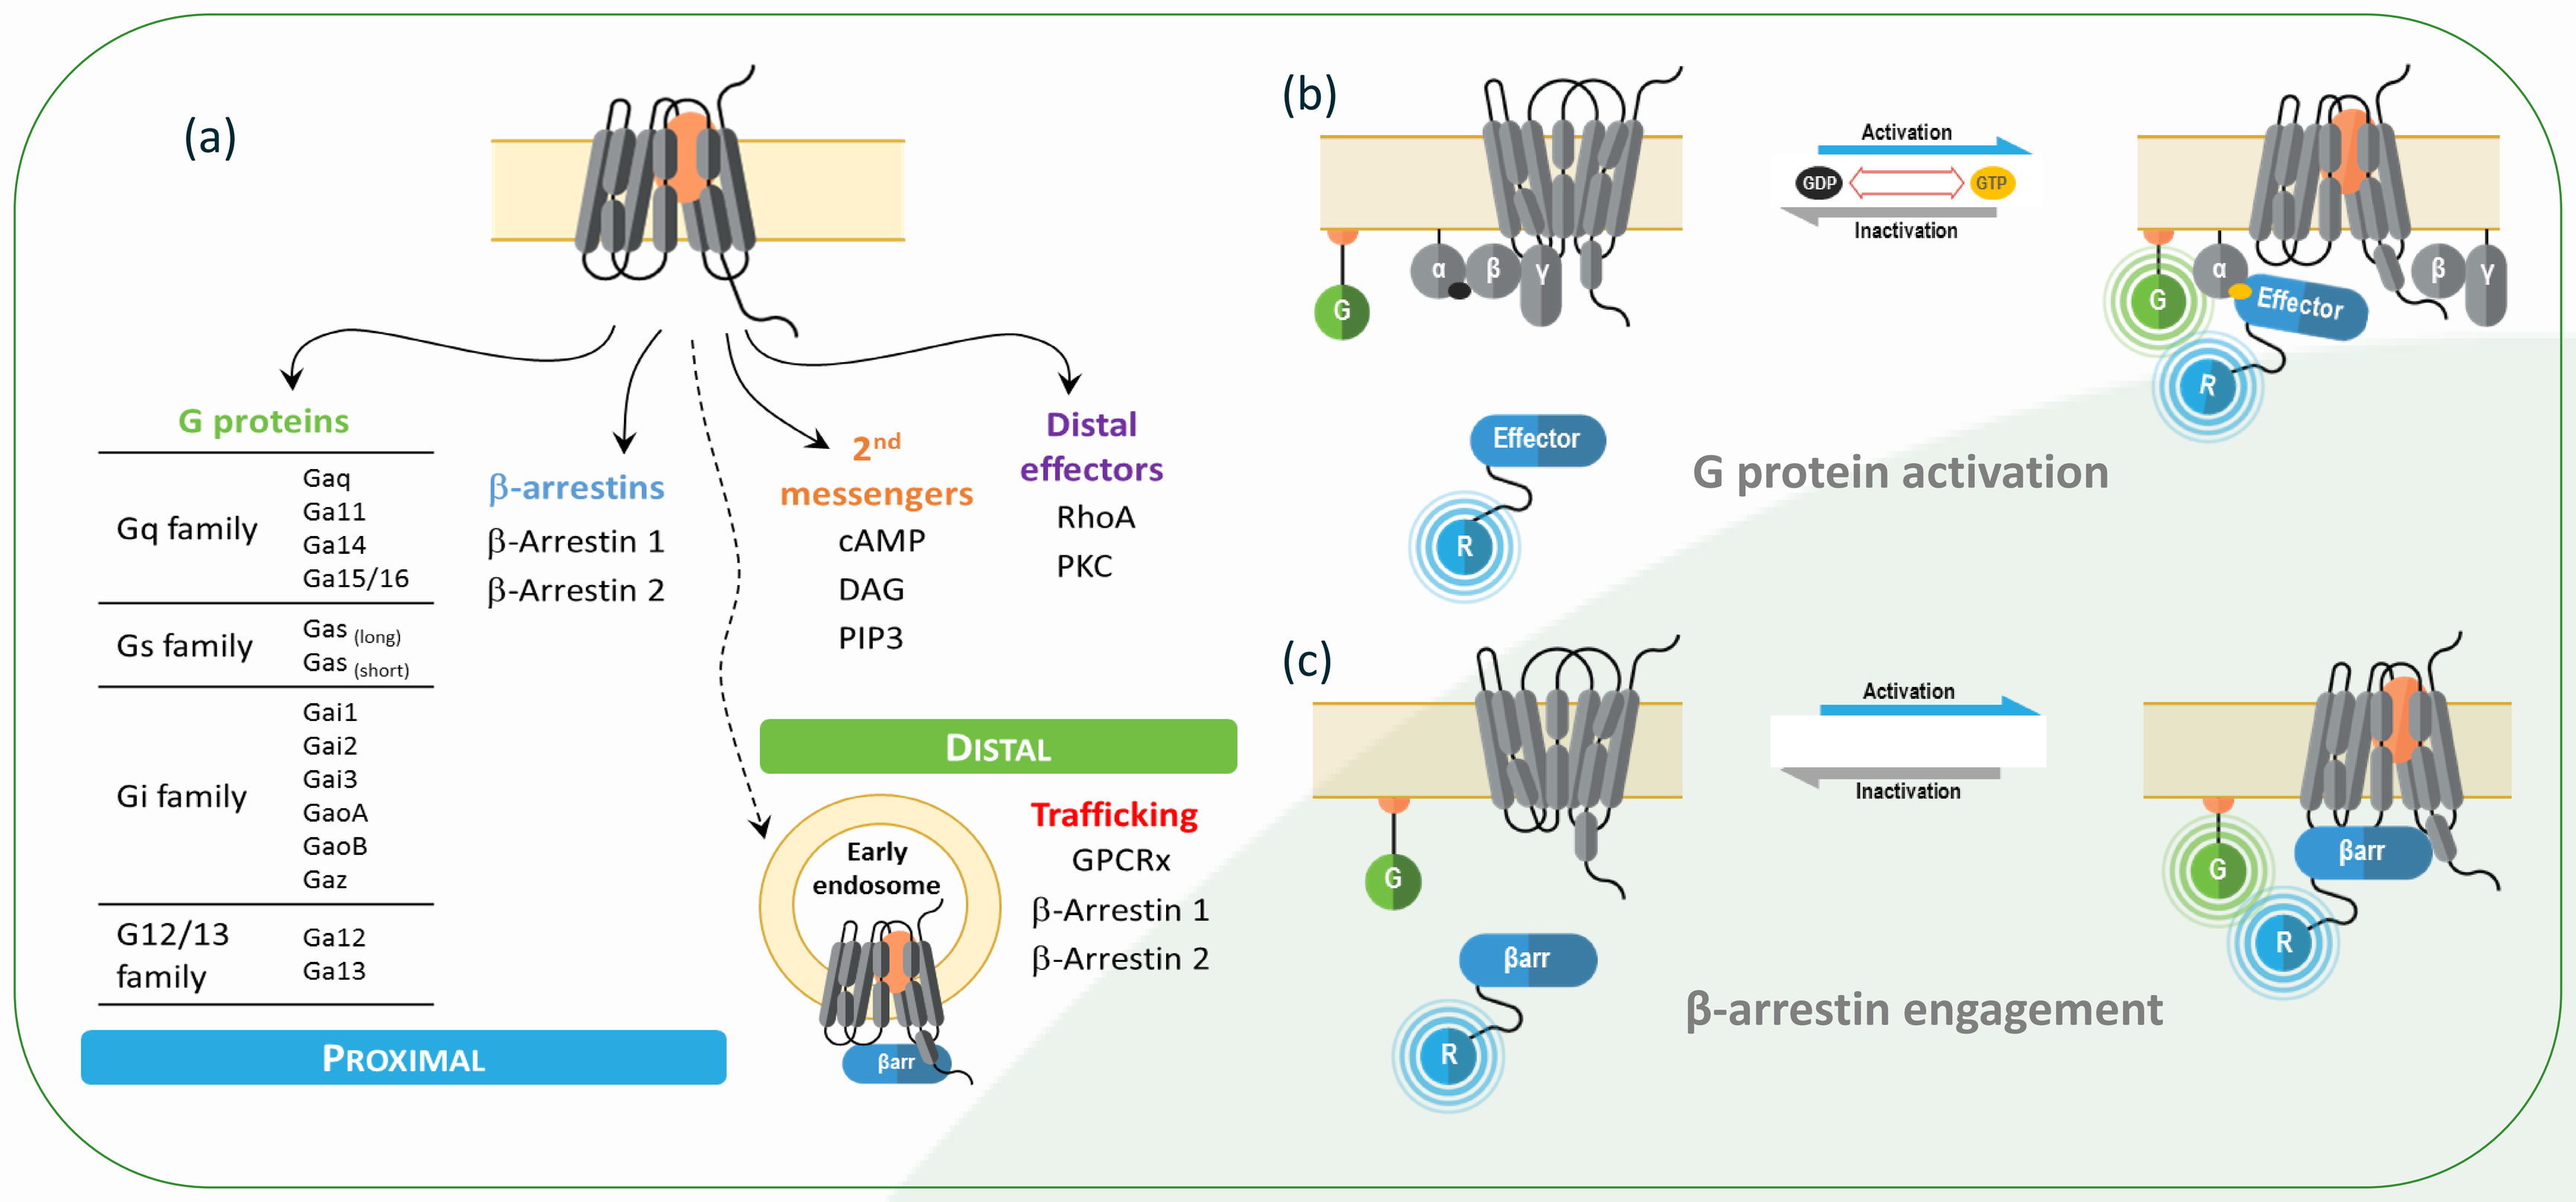
(a)
G proteins
Gq family
Gaq
Ga11
Ga14
Ga15/16
Gs family
Gas (long)
Gas (short)
Gi family
Gai1
Gai2
Gai3
GaoA
GaoB
Gaz
G12/13 family
Ga12
Ga13
β-arrestins
β-Arrestin 1
β-Arrestin 2
2nd messengers
cAMP
DAG
PIP3
Distal effectors
RhoA
PKC
DISTAL
Early endosome
Trafficking
GPCRx
β-Arrestin 1
β-Arrestin 2
PROXIMAL
(b)
Activation
Inactivation
GDP
GTP
Effector
G protein activation
(c)
Activation
Inactivation
βarr
β-arrestin engagement

Fig. 1. BRET-based biosensors for G protein and βarr activation. (a) The bioSens-All® platform consists of 16 biosensors for monitoring activation of specific Gα proteins and βarrs downstream of GPCRs. These biosensors measure receptor proximal events engaged upon receptor activation, namely G protein activation and βarr recruitment. In addition to proximal biosensors, the technology includes sensors detecting distal effector activity and second messenger generation. **(b)** Upon receptor activation, RlucII-tagged effector proteins (Effector-RlucII) translocate towards and interact with active Gα subunits from each G protein family, leading to increased BRET. **(c)** Upon receptor activation, RlucII-tagged βarrs (βarr-RlucII) translocate to the plasma membrane, thus increasing BRET with membrane-anchored rGFP.

Automated Screening Platform

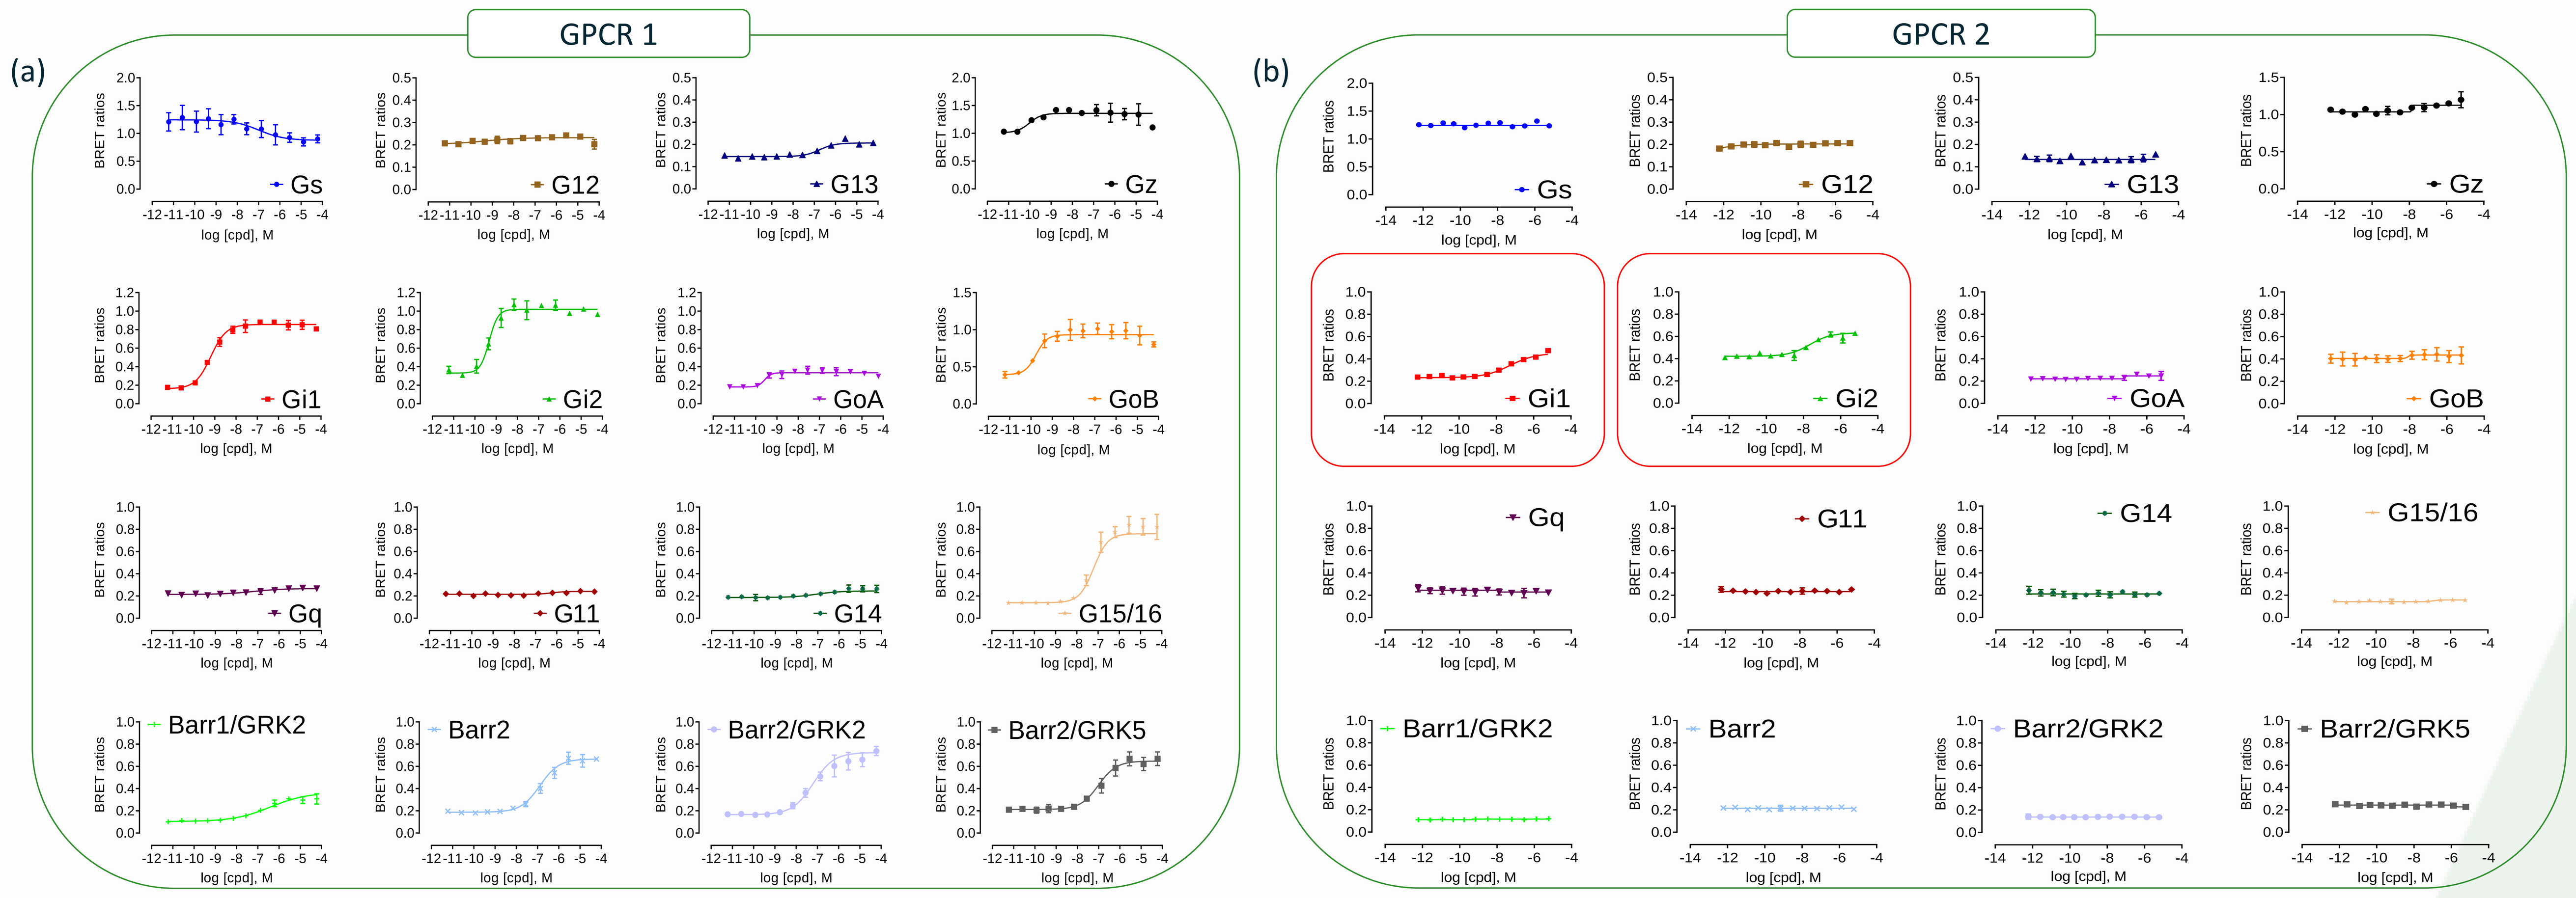
GPCR 1
GPCR 2
(a)
(b)
Gs
G12
G13
Gz
Gi1
Gi2
GoA
GoB
Gq
G11
G14
G15/16
Barr1/GRK2
Barr2
Barr2/GRK2
Barr2/GRK5
BRET ratios
log [cpd], M

Fig. 2. Signaling profiles of WT human GPCRs. HEK293 cells were transfected with plasmids coding for a GPCR and one of 16 biosensors. Cells were treated with GPCR-specific ligand and response was recorded 30 mins later. **(a)** GPCR 1 demonstrated promiscuous signaling across many G protein and βarr pathways. **(b)** GPCR 2 demonstrated selective signaling on Gi1 and Gi2 pathways only.

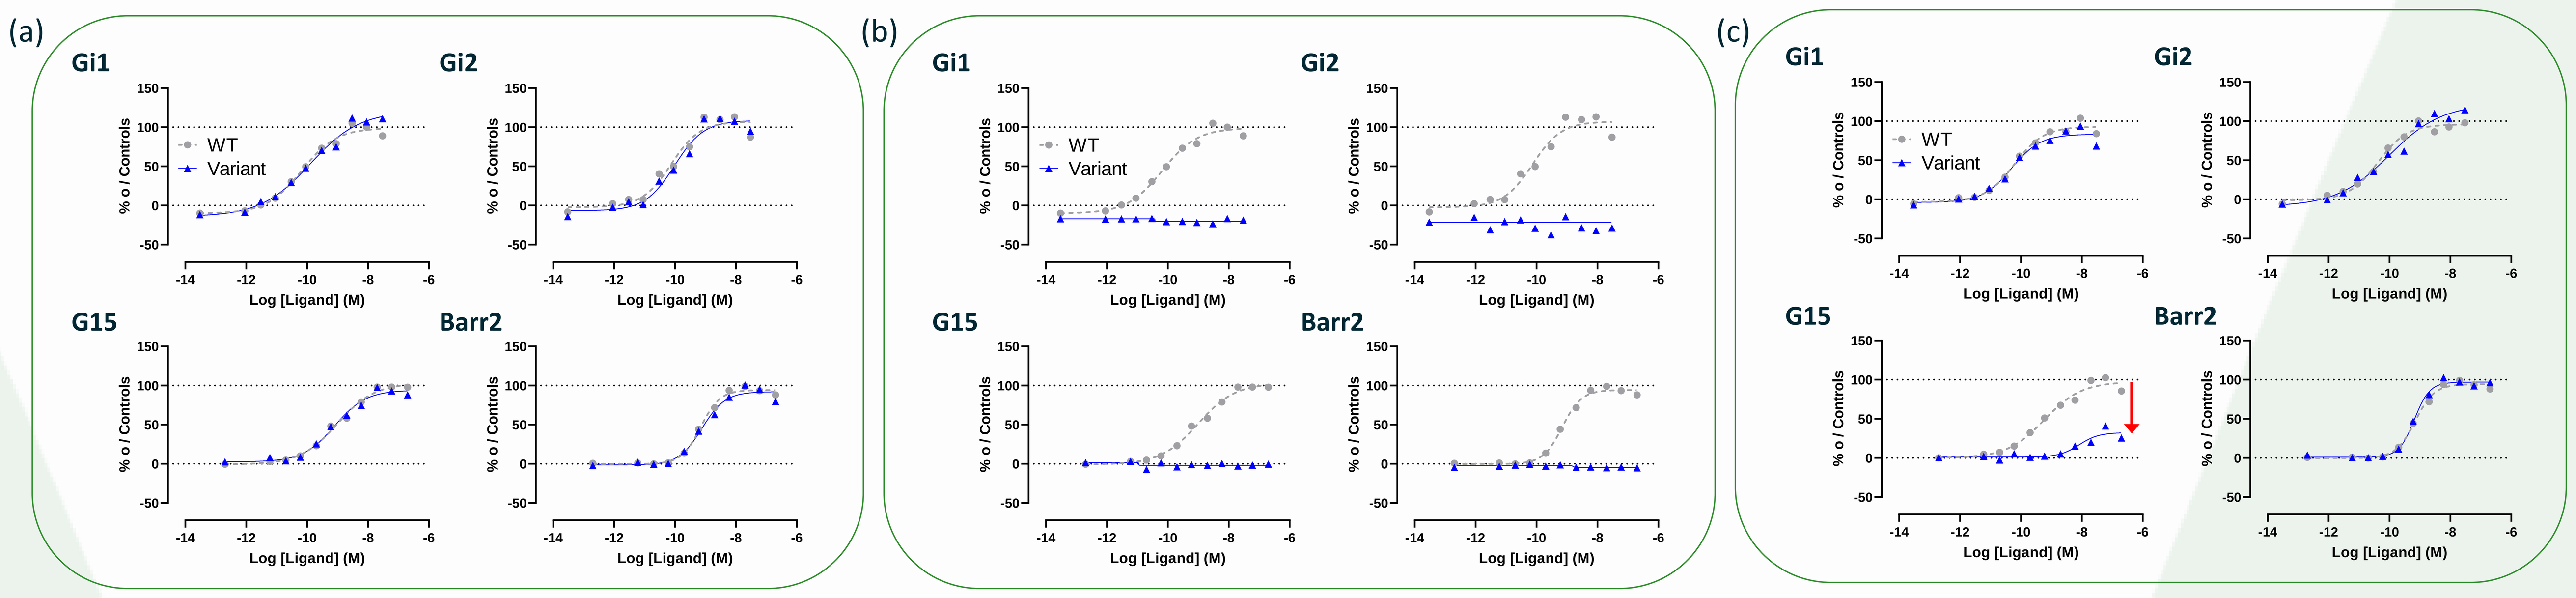
(a)
(b)
(c)
Gi1
Gi2
G15
Barr2
WT
Variant
% o / Controls
Log [Ligand] (M)

Fig. 3. Signaling profiles of variant receptors. HEK293 cells were transfected with plasmids coding for a specific GPCR and one of 16 biosensors. Cells were treated with GPCR-specific ligand and response recorded 30 mins later. **(a)** Variant displays WT-like profile on all pathways selected. **(b)** Variant displays complete loss of function profile on all pathways selected. **(c)** variant displays loss of function on G15 and WT-like profile on Gi1, Gi2, and βarr2, thus yielding an overall biased profile.

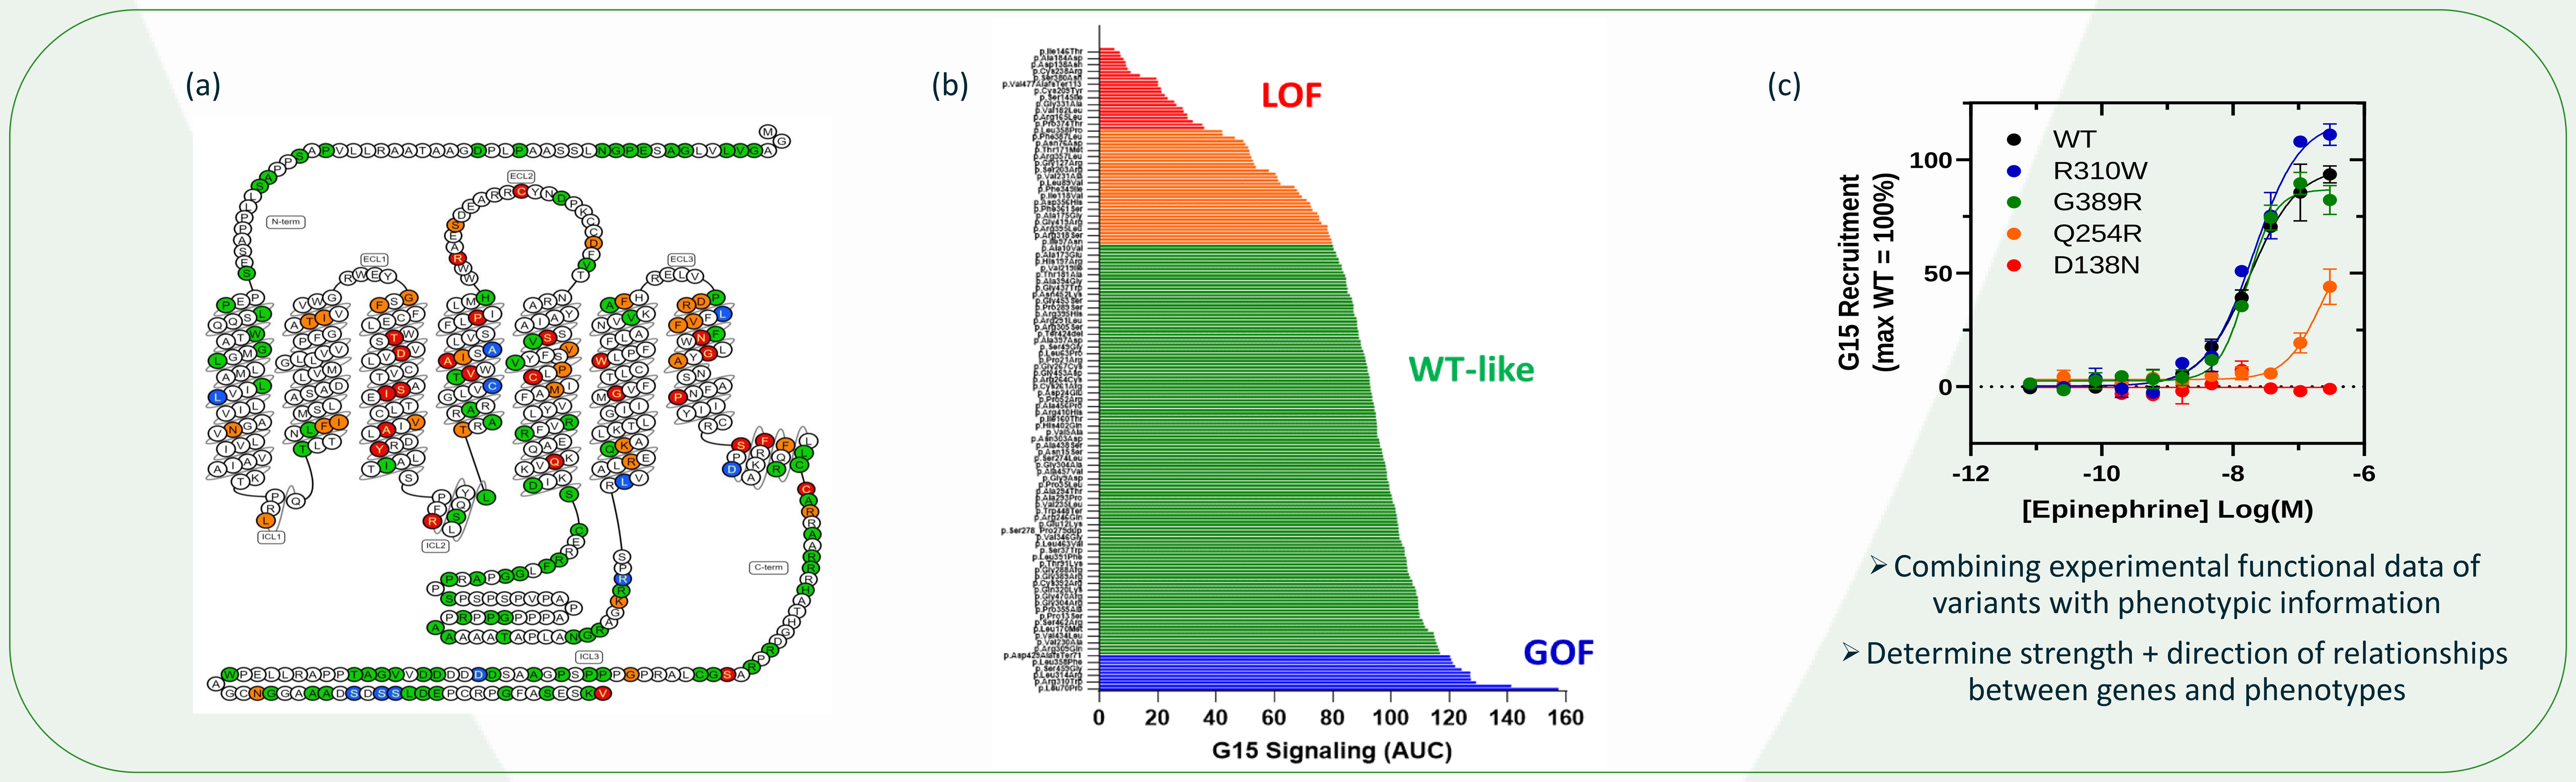
(a)
N-term
ECL1
ECL2
ECL3
ICL1
ICL2
ICL3
C-term
(b)
LOF
WT-like
GOF
G15 Signaling (AUC)
(c)
WT
R310W
G389R
Q254R
D138N
G15 Recruitment (max WT = 100%)
[Epinephrine] Log(M)

- Combining experimental functional data of variants with phenotypic information
- Determine strength + direction of relationships between genes and phenotypes

Fig. 4. Activity of 265 variants of ADRB1. (a) Location of variants on the serpentine map of ADRB1. **(b)** In vitro pharmacology of G15 area under the curve (AUC) for all profiled ADRB1 variants. **(c)** G15 signaling of four representative variants.

Mechano-Stimulation Assay

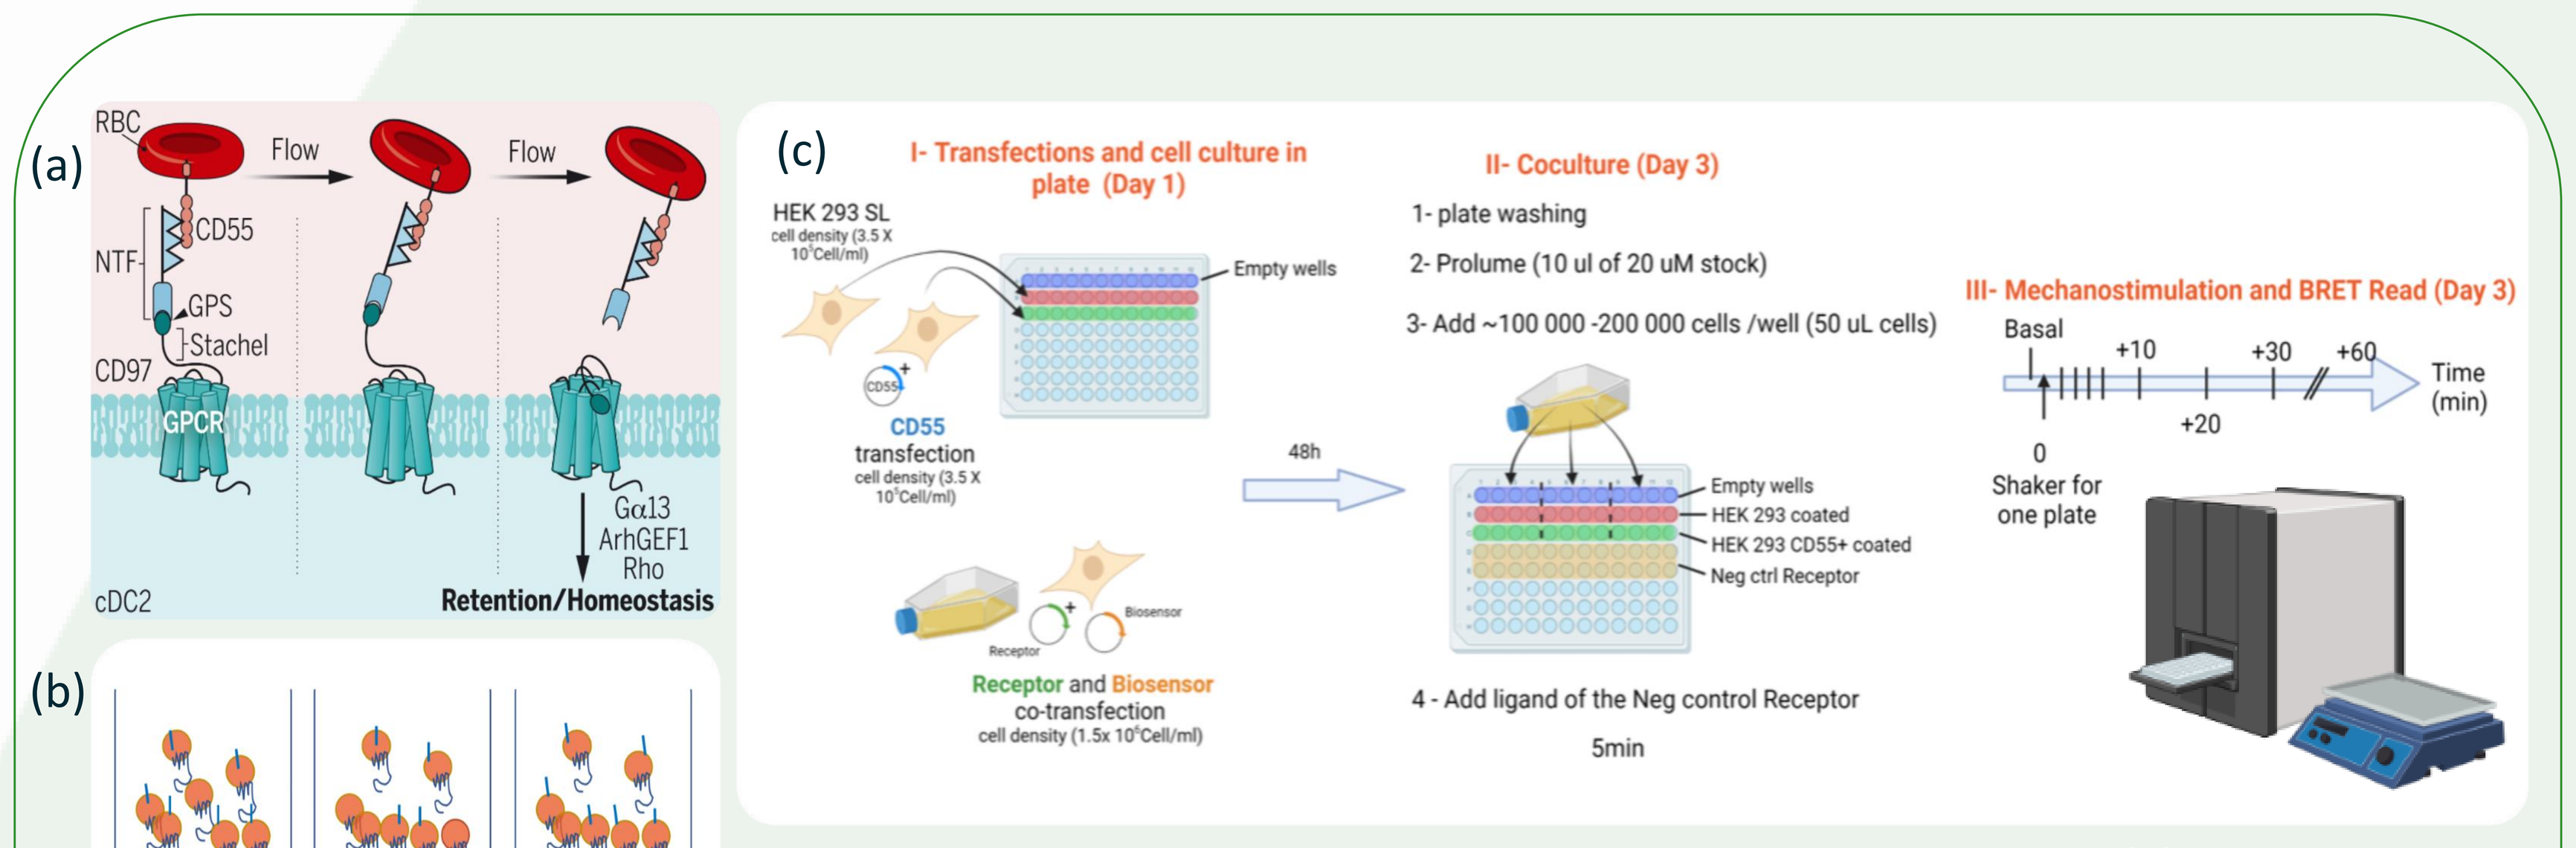
(a)
RBC
Flow
Flow
CD55
NTF
GPS
Stachel
CD97
GPCR
cDC2
Gα13
ArhGEF1
Rho
Retention/Homeostasis
(b)
Empty Wells
Adhered HEK-293
Adhered CD55+ HEK-293
HEK-293
ADGRE5
CD55
Other ligands?
(c)
I- Transfections and cell culture in plate (Day 1)
HEK 293 SL
CD55 transfection
Receptor and Biosensor co-transfection
48h
II- Coculture (Day 3)
1- plate washing
2- Prolume (10 ul of 20 uM stock)
3- Add ~100 000 -200 000 cells /well (50 uL cells)
Empty wells
HEK 293 coated
HEK 293 CD55+ coated
Neg ctrl Receptor
4 - Add ligand of the Neg control Receptor
5min
III- Mechanostimulation and BRET Read (Day 3)
Basal
Shaker for one plate
Time (min)

Fig. 5. Schematic representation of mechano-stimulation workflow. (a) Illustration of adhesion GPCR activation by Dan Liu. et al, Science (2022). **(b and c)** HEK293 cells co-transfected with hADGRE5 and biosensor were seeded into empty wells or pre-coated wells with HEK293 in 96-wells plates and subjected or not to vigorous orbital shaking before BRET measurements.

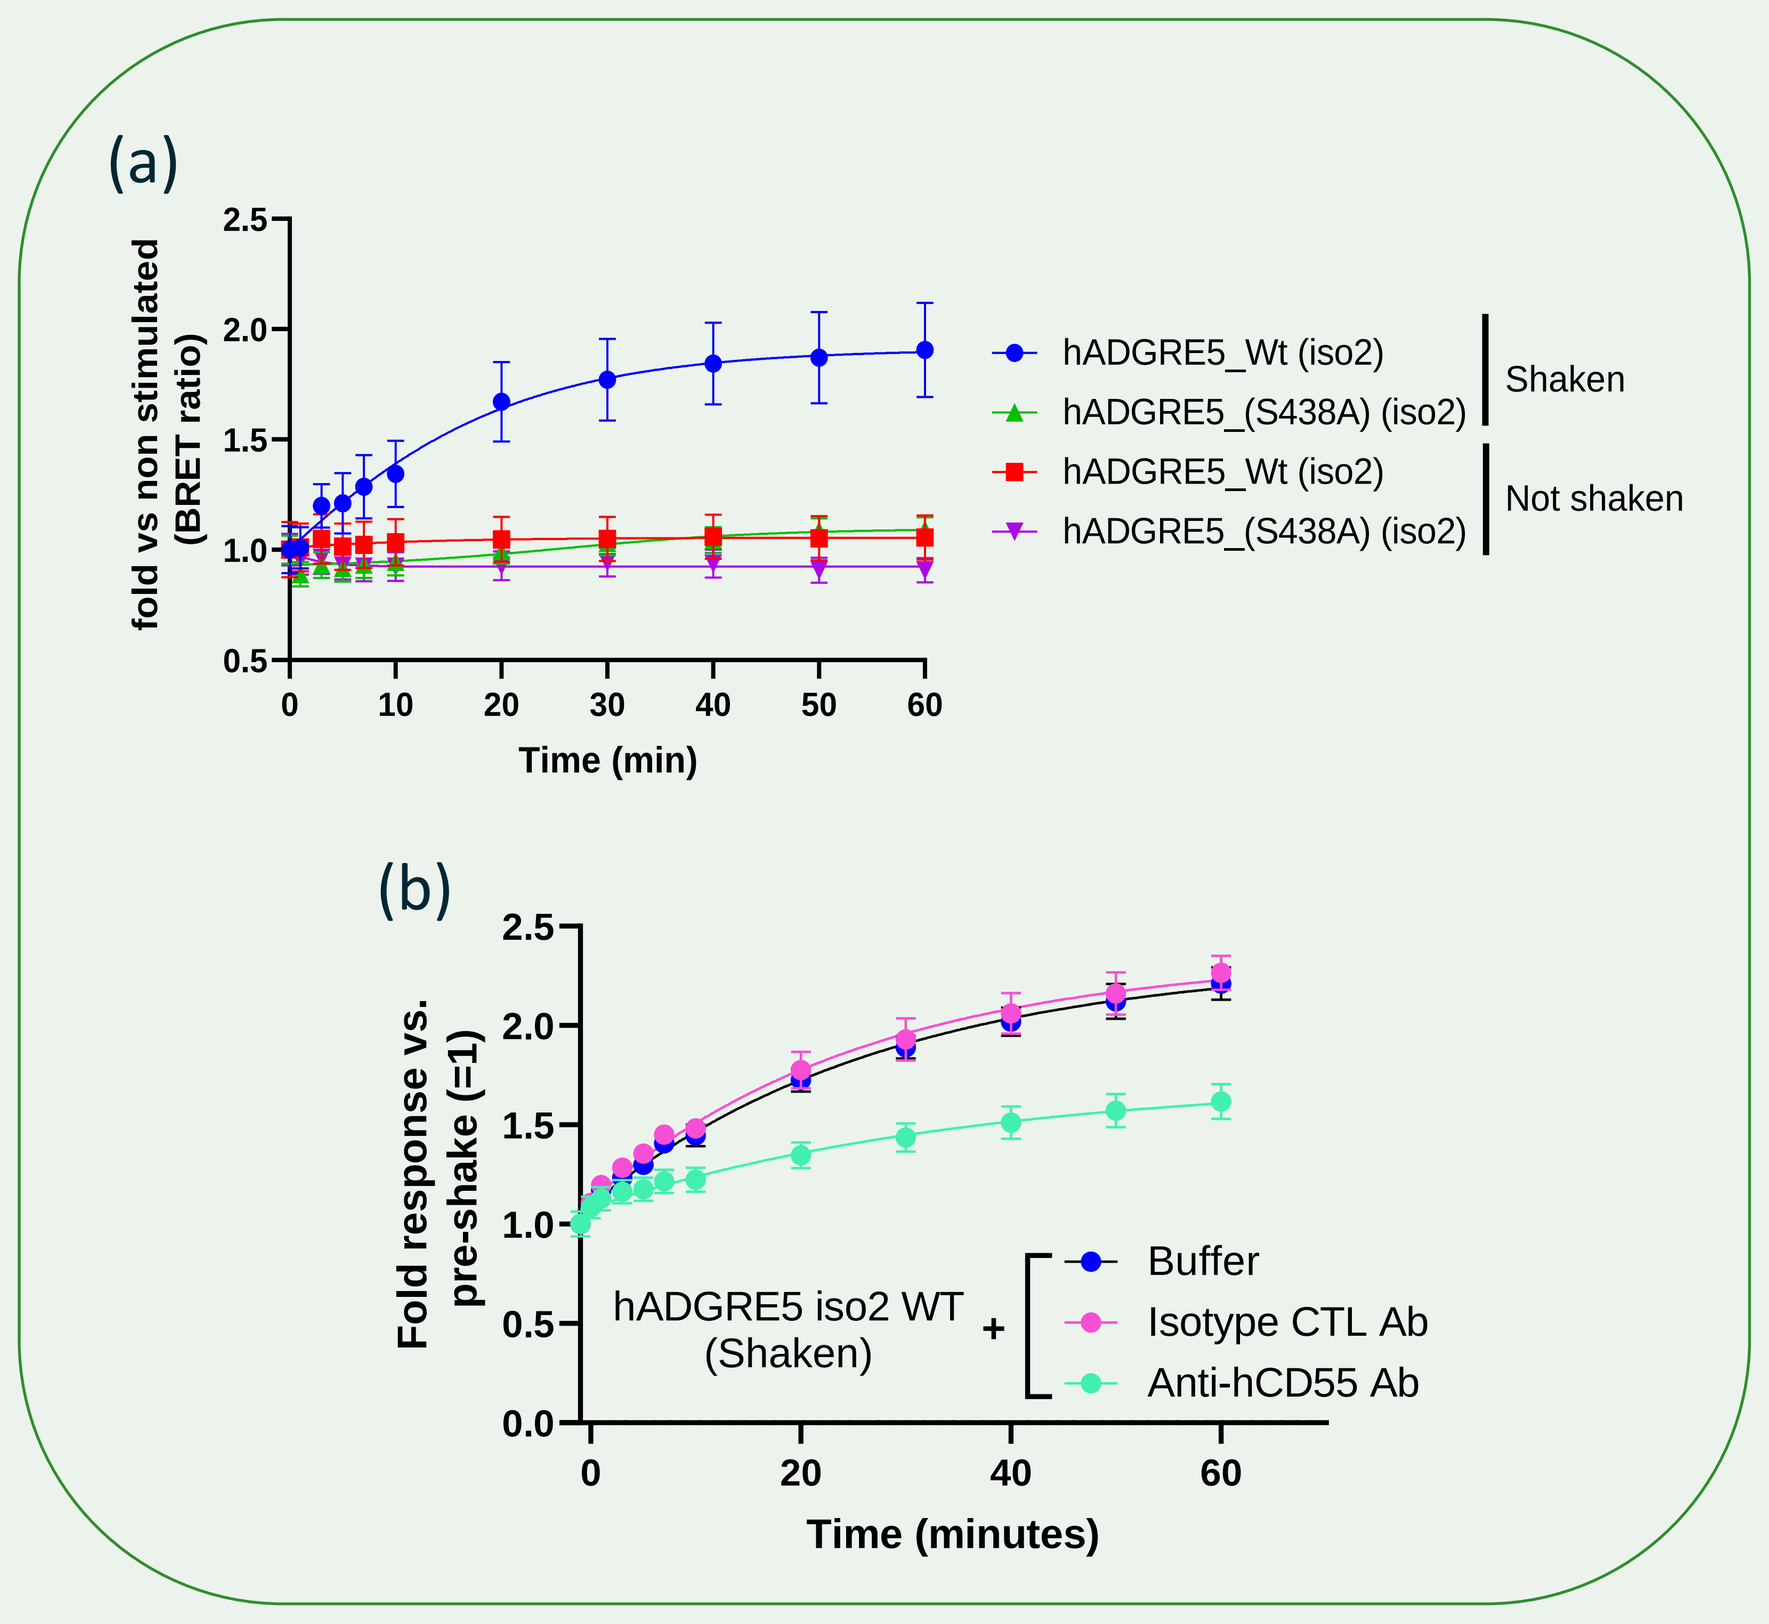
(a)
fold vs non stimulated (BRET ratio)
Time (min)
hADGRE5_Wt (iso2)
hADGRE5_(S438A) (iso2)
Shaken
hADGRE5_Wt (iso2)
hADGRE5_(S438A) (iso2)
Not shaken
(b)
Fold response vs. pre-shake (=1)
Time (minutes)
hADGRE5 iso2 WT (Shaken)
Buffer
Isotype CTL Ab
Anti-hCD55 Ab

Fig. 6. Measuring mechano-stimulation of hADGRE5 through β-arr plasma membrane recruitment sensor. (a and b) HEK293 cells co-transfected with biosensor and hADGRE5 or uncleavable GPS point mutant S438A-hADGRE5 were seeded into pre-coated wells with HEK293 in 96-wells plates and subjected or not to vigorous orbital shaking before BRET measurements. **(b)** Cells were incubated with neutralizing anti-CD55 antibody, isotype control antibody, or buffer.

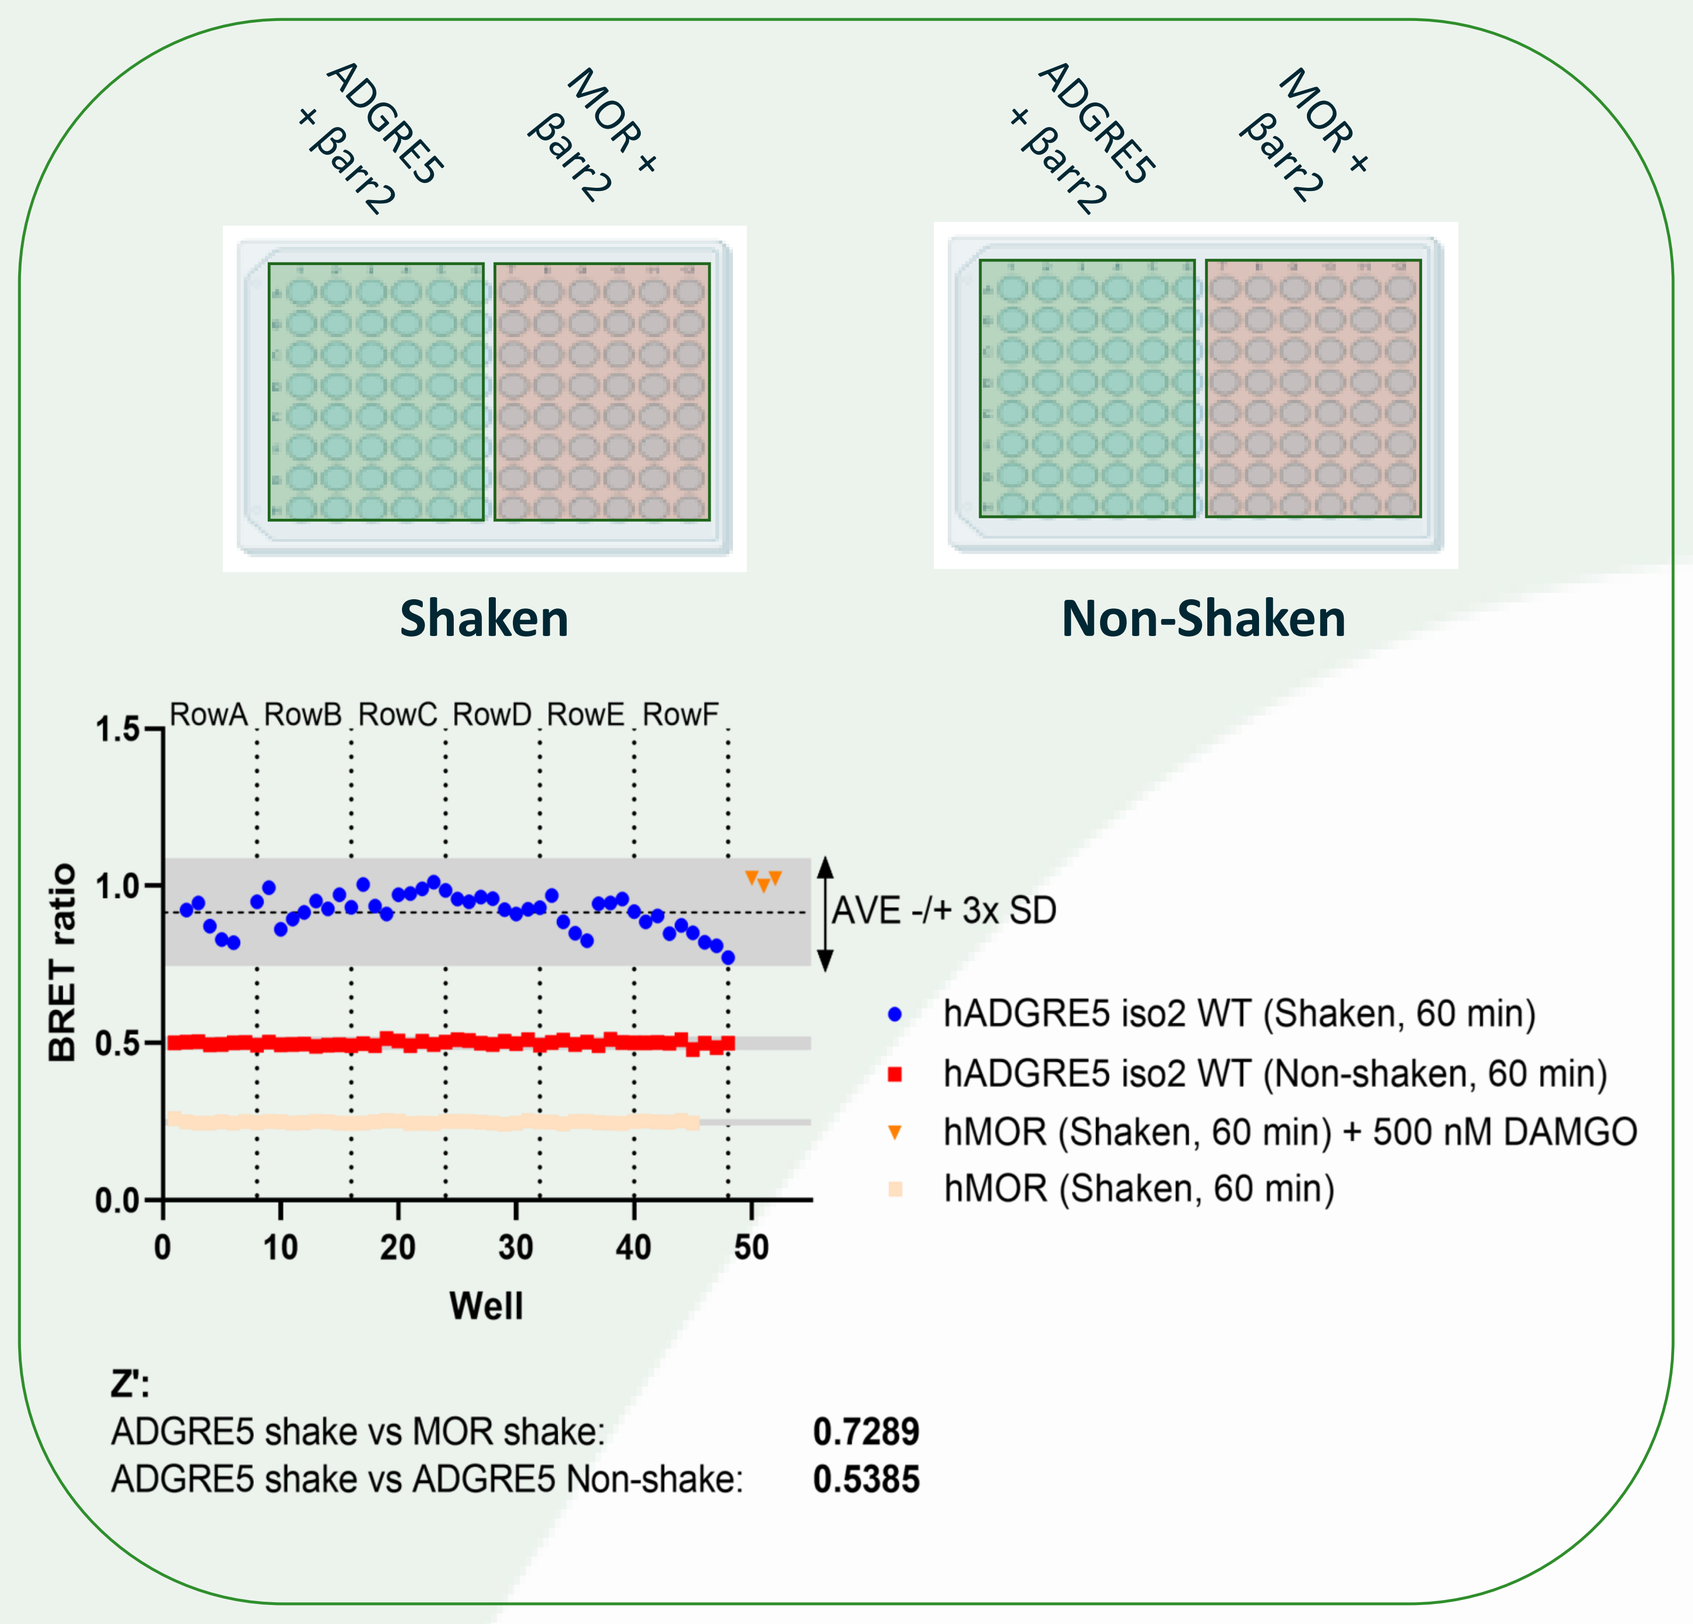
ADGRE5 + Barr2
MOR + Barr2
Shaken
Non-Shaken
RowA RowB RowC RowD RowE RowF
AVE -/+ 3x SD
BRET ratio
Well
hADGRE5 iso2 WT (Shaken, 60 min)
hADGRE5 iso2 WT (Non-shaken, 60 min)
hMOR (Shaken, 60 min) + 500 nM DAMGO
hMOR (Shaken, 60 min)
Z':
ADGRE5 shake vs MOR shake: 0.7289
ADGRE5 shake vs ADGRE5 Non-shake: 0.5385

Fig. 7. Z' robustness of mechano-stimulation BRET assay. HEK293 cells co-transfected with β-arr plasma membrane recruitment sensor and hADGRE5 or MOR were subjected or not to vigorous orbital shaking before BRET measurements. Each point represents the BRET ratio value from one well belonging to the row indicated between the dotted lines.

Conclusions

- bioSens-All® is a robust and flexible platform to assess GPCR signaling:
 - Uses unmodified receptors and G proteins
 - Permits pan-pathway analysis in parallel using one protocol
 - Automation of platform allows large scale GPCR variant profiling
 - Adaptable to assess physiologically relevant modes of adhesion GPCR activation
- Pairing signaling data generated through bioSens-All® with clinical phenotypes can reveal associations between altered receptor pharmacology and disease states
- Facilitate the validation of novel therapeutic targets and discovery of innovative therapeutic agents